A PHASE 2 STUDY OF ORAL MKC-1 FOR METASTATIC BREAST CANCER (MBC)

B.P. Schneider[1], M. Karwal[2] , L. Laufman[3], L. Sylvester[4], M.A. Taylor[5], C.F. Sidor[6], A.L. Hannah[7], J. Arnott[6], K.D. Miller[1]

[1]Indiana University, Indianapolis, IN; [2]University of Iowa, Iowa City, IA; [3]Hematology Oncology Consultants, Inc., Columbus, OH; [4]Integrated Community Oncology Associates, Jacksonville, FL; [5]St. Joseph's/Candler Health System, Inc., Savannah, GA; [6]EntreMed, Inc., Rockville, MD; [7]Consultant, Sebastopol, CA,

ABSTRACT

Introduction: MKC-1 is a novel oral cell cycle inhibitor with *in vitro* and *in vivo* activity against many tumor cell lines, including breast cancer. Proteins identified as binding targets of MKC-1 include microtubules and members of the importin-β family. Additionally, MKC-1 has demonstrated an ability to inhibit the Akt-mTOR signaling pathway *in vitro* and *in vivo*. This phase 2 study evaluated the efficacy and safety of MKC-1.

Methods: Eligible patients (pts) had MBC previously treated with an anthracycline (A) and a taxane (T). Pts with treated/stable CNS metastases were eligible. Pts were treated with MKC-1 at 125 mg/m² BID D1-14 of every 28 day cycle. Dose escalations/reductions were made based on first cycle neutropenia. The primary endpoint was objective response rate (ORR) by RECIST. Enrollment was to continue to 53 evaluable pts if ≥2 of the first 23 responded.

Results: A total of 65 pts have now been enrolled. Median age/ECOG performance status was 59/0. Receptor status: 52%/41% ER/PR+ and 26% Her2+. Median number of cycles was 2 (range 1-8). Grade ≥3 drug-related toxicity (n=58) was as follows: elevated LFTs (9%), sensory neuropathy (2%), elevated creatinine (2%), mucositis (2%), and neutropenia (8%), including two deaths from neutropenic sepsis. 5 pts discontinued due to toxicity. Of pts proceeding to cycle 2, 57% and 10% had the dose increased or reduced, respectively. 55 patients were evaluable for response. ORR was 3/55 (5.5%) and an additional 5 pts had stable disease. 1 pt (adjuvant A/cyclophosphamide/paclitaxel and docetaxel/cisplatin for metastatic disease) had a PR (observed after cycle 4; confirmed after cycle 6; withdrawal for progression at cycle 8). The 2nd pt (adjuvant CMF with A/docetaxel, capecitabine and nab-paclitaxel for metastatic disease) had an unconfirmed CR after 3 cycles. Confirmatory scans were not possible because pt chose to withdraw from study. The 3rd pt (AC/docetaxel for metastatic disease) had a PR and remained on study for 5 cycles.

Conclusions: MKC-1 is well tolerated at its recommended dose and schedule. Activity is observed in pts previously receiving A/T for MBC.

NONCLINICAL DATA

Figure 1. MKC-1 Induces Aberrant Spindle Formation and Disrupts Microtubules

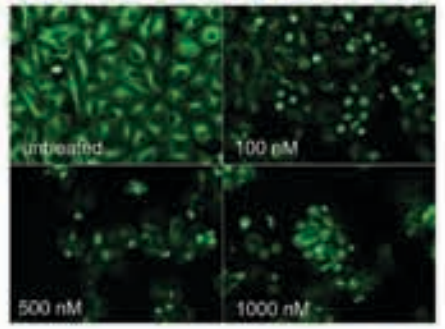
untreated
100 nM
500 nM
1000 nM

Figure 2. Dosing Twice Daily (at Equivalent Total Daily Dose) Produces Better Efficacy at a More Tolerable Dose. In the MDA-MB-435 model of human breast cancer cells, MKC-1 (50 mg/kg BID) induces significant inhibition of tumor growth with continued body weight gain in mice, whereas 100 mg/kg as a single dose is lethal.

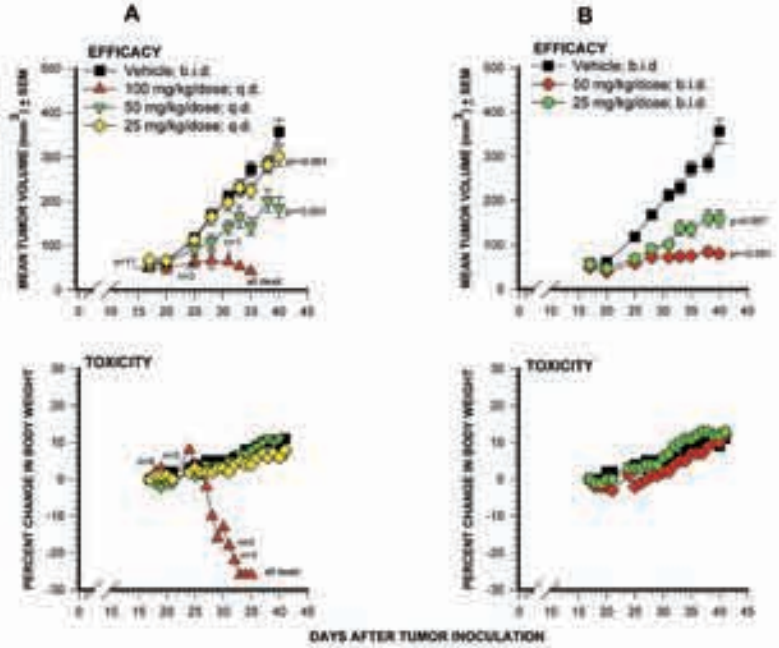
A
B
EFFICACY
TOXICITY
DAYS AFTER TUMOR INOCULATION

Figure 3. MKC-1 Inhibits the Akt and mTOR Signaling Pathways in the MDA-MB-231 Breast Cancer Cell Line

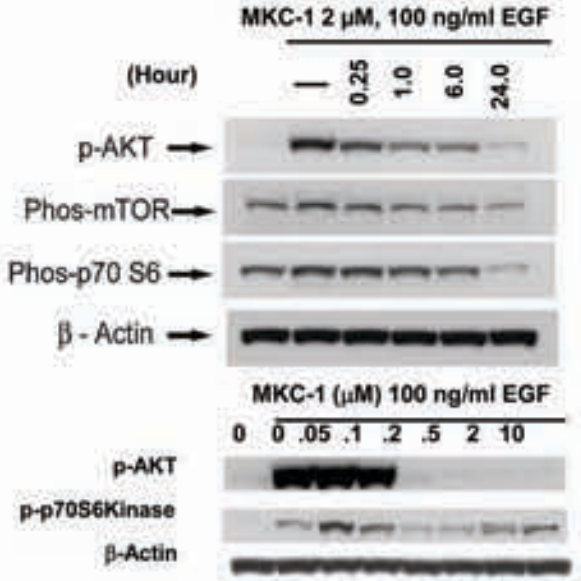
MKC-1 2 μM, 100 ng/ml EGF
(Hour)
0.25
1.0
6.0
24.0
p-AKT
Phos-mTOR
Phos-p70 S6
β - Actin
MKC-1 (μM) 100 ng/ml EGF
0 0 .05 .1 .2 .5 2 10
p-AKT
p-p70S6Kinase
β-Actin

STUDY DESIGN

- Patients received oral MKC-1 daily for 14 days followed by a 14-day break in therapy.
- Patients underwent weekly safety monitoring Cycle 1 (decreased to every 2 weeks in Cycle 2 and beyond) with efficacy assessments every 8 weeks.
- Primary efficacy endpoint: RECIST confirmed objective response rate (either PR or CR)
- Simon optimal two-stage design (boundaries <5%, >17%)
 Stage 1: at least 2 responses required in 23 evaluable patients to move to the 2nd stage
 Stage 2: at least 5 confirmed responses in 53 evaluable patients to reject the null hypothesis

(MKC-1 does not have less than a <5% ORR; type I and II errors of 10%, respectively).

PHASE 2 CLINICAL DATA

Enrollment	Patients (n=65)
Gender	All female
Median Age/ECOG PS	59 / 0
Receptor Status at Diagnosis *	52%ER+ 41% PR+ 26% Her2+
Median number of cycles (range)	2 (1-8)
Prior Anthracycline Neo/Adjuvant* Metastatic * *	 81% 27%
Prior Taxane Neo/Adjuvant* Metastatic **	 70% 50%
Median Number of Prior Chemotherapy Regimens in the Metastatic Setting (range)***	2 (1-8)

* data based on 57 pts; ** data based on 56 pts; *** data based on 53 pts

Response	Evaluable Patients (n=55)
Complete Response	1 (unconfirmed)
Partial Response	2 (1 confirmed)
Stable Disease >4 months	5

One patient had a reduction in cutaneous disease while on the initial 14-day cycle at 125 mg/m² BID. Dose escalation to 150 mg/m² BID markedly improved her cutaneous disease but resulted in severe toxicity, including neutropenic fevers. Although continuous dosing was attempted at 100 mg BID, the patient showed signs of progressive disease at this tolerable dose.

PATIENT #38005: CONFIRMED PR

Baseline 14 Mar 07
Right axillary LN on CT

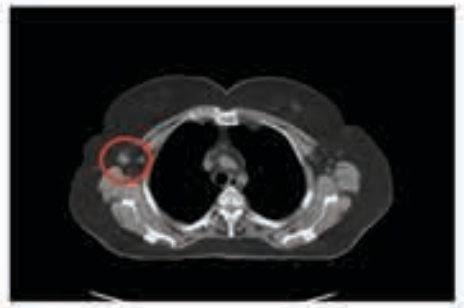

Best response 07 Jun 07
Right axillary LN on CT

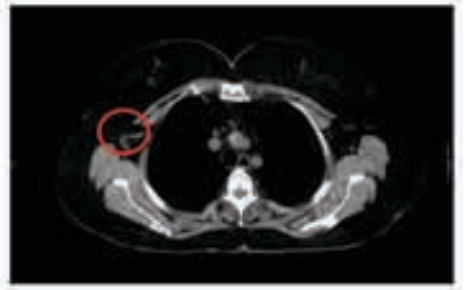

Observed Percent Change in the Sum of Tumor Sizes from Baseline to the Minimum Post-Baseline Sum

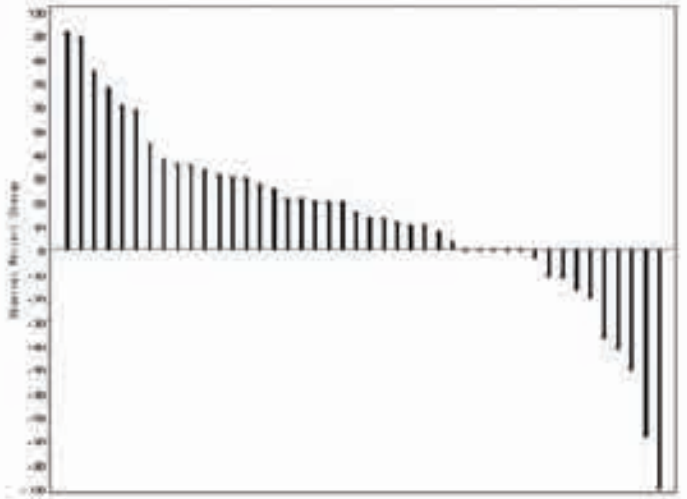

SAFETY DATA (all toxicity grades) n=58

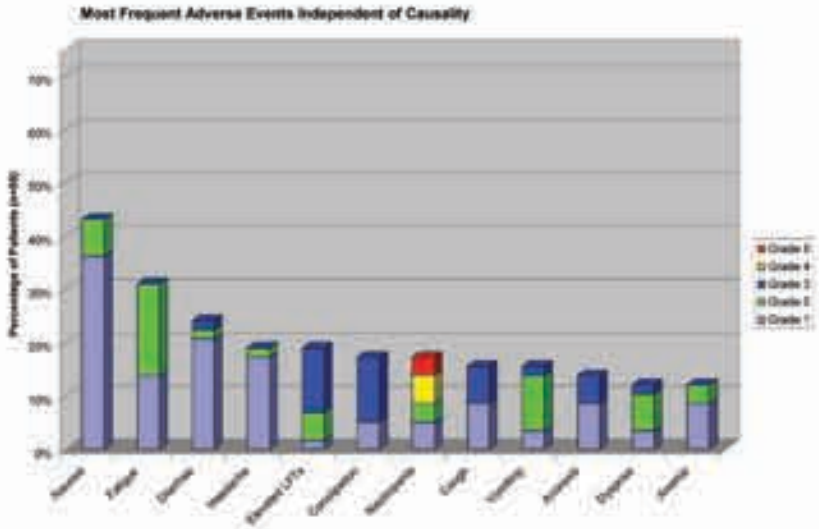
Most Frequent Adverse Events Independent of Causality

CONCLUSIONS

- Although there was antitumor activity in this highly refractory breast cancer population, MKC-1 did not meet the pre-specified goal of 5 confirmed responses.
- Common toxicities included gastrointestinal (N/V/D with constipation), myelosuppression (neutropenia with anemia), fatigue, and headache.
- Neutropenia correlated with baseline serum albumin, likely due to a change in the ratio of free/bound drug. Patients with severe pre-existing hypoalbuminemia are at increased risk of severe toxicity, including neutropenia and mucositis.
- Plans are underway to study MKC-1 in less refractory, enriched populations in metastatic breast cancer (based on its known molecular targets, such as HIF-1α or mTOR).

Structure of MKC-1

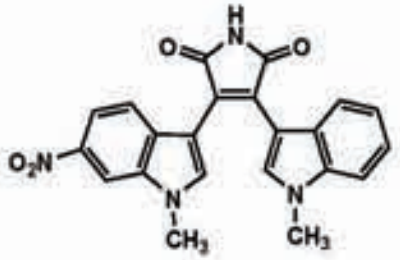

3-(1-Methyl-1H-indol-3yl)-4-(1-Methyl-6-nitro-1H-indol-3-yl)-1H-pyrrole-2,5-dione

Miikana Therapeutics
An EntreMed, Inc. Company

ENTREMED