The Novel Aurora Kinase Inhibitor ENMD-2076 Has Potent Activity against Multiple Myeloma (MM) *in Vitro* and *in Vivo*, and Shows Synergistic Activity in Combination with Lenalidomide

Xiaojing Wang[1], Anthony L. Sinn[2], Attaya Suvannasankha[1], Colin D. Crean[1], Li Chen[1], Shuhong Zhang[1], Jing Liang[1], Genglin Zhang[1], Karen E. Pollok[2], Rafat Abonour[1], Carolyn Sidor[3], Mark R. Bray[3], Sherif S. Farag[1]

[1]Department of Medicine , Division of Hematology and Oncology, Indiana University School of Medicine; [2]Indiana University Simon Cancer Center, *In Vivo* Therapeutics Core, Indianapolis, Indiana; and [3]EntreMed, Inc., Toronto, Ontario, Canada

MELVIN AND BREN SIMON CANCER CENTER
INDIANA UNIVERSITY

INDIANA UNIVERSITY
Center for IMMUNOBIOLOGY

ABSTRACT

ENMD-2076 is a novel, orally-active molecule that has been shown to have significant activity against Aurora A kinase as well as multiple receptor tyrosine kinases (RTK). We investigated the single agent activity of ENMD-2076 against MM cells *in vitro* and *in vivo*, and in combination with lenalidomide. ENMD-2076 free base showed significant cytotoxicity against MM cells with a mean LC50 of 3.84±0.86 µM at 48 hours *in vitro*. Cytotoxicity was associated with cleavage of caspase 3, 8, 9 and PARP, and loss of mitochondrial membrane potential as early as 6 hours. ENMD-2076 free base inhibited downstream targets phosphorylated (p)-BAD, p-Foxo1a and p-GSK-3β at 6 hours and inhibition of Aurora kinases was observed at 24 hours. NOD/SCID mice implanted with H929 human plasmacytoma xenografts and treated for 30 days with 50, 100, 200mg/kg/d ENMD-2076 showed a dose-dependent inhibition of tumor growth, with minimal toxicity as assessed by the stable weight of treated animals. Immunohistochemical staining of tumors from sacrificed animals showed significant reduction in Ki67, p-H3 and CD34, and an increase in caspase-3 at all dose levels of treatment compared to control tumors. A reduction in p-FGFR-3 was observed on Western blot from the lysates of H929 tumors obtained from treated animals. ENMD-2076 free base also showed synergistic cytotoxic activity when combined with lenalidomide against H929, MM1.R and MM1.S cells as assessed by MTT assay and Annexin-V/PI staining. Using the Chou-Talalay method, the combination indices (CI) were < 1 for all three cell lines across a range of concentrations of ENMD-2076 free base (0.25-1.0 µM) plus lenalidomide (2.5-10 µM) indicating synergistic activity (CI=0.362 H929; CI=0.315 MM1.R; CI=0.415 MM1.S). Our results provide rationale for the investigation of ENMD-2076 alone and in combination with lenalidomide in patients with multiple myeloma.

METHODS

Cytotoxicity was assessed using MTT assay and annexin V/PI staining, and cell apoptosis and cell cycle were analyzed by flow cytometry, and intracellular signaling by Western blotting. Human plasmacytoma xenograft mouse model was set up in NOD-SCID mice using human MM cell line NCI-H929.

RESULTS

1. ENMD-2076 is cytotoxic to MM cells.

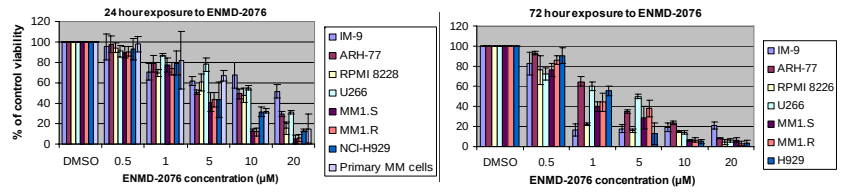
24 hour exposure to ENMD-2076
72 hour exposure to ENMD-2076
% of control viability
ENMD-2076 concentration (µM)
DMSO
0.5
1
5
10
20
IM-9
ARH-77
RPMI 8228
U266
MM1.S
MM1.R
NCI-H929
Primary MM cells
H929

2. ENMD-2076 induces apoptosis of MM cells *in vitro* with activation of caspases.

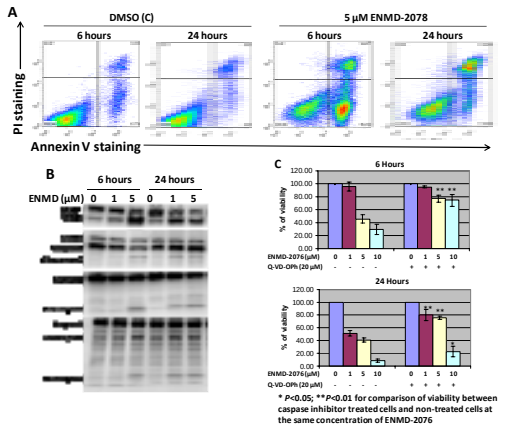
DMSO (C)
5 µM ENMD-2078
6 hours
24 hours
PI staining
Annexin V staining
6 Hours
24 Hours
% of viability
* P<0.05; **P<0.01 for comparison of viability between caspase inhibitor treated cells and non-treated cells at the same concentration of ENMD-2076

3. ENMD-2076 cleaves Mcl-1 and downregulates pro-apoptotic proteins XIAP and survivin, and inhibits the PI3K/Akt pathway with downregulation of downstream effectors.

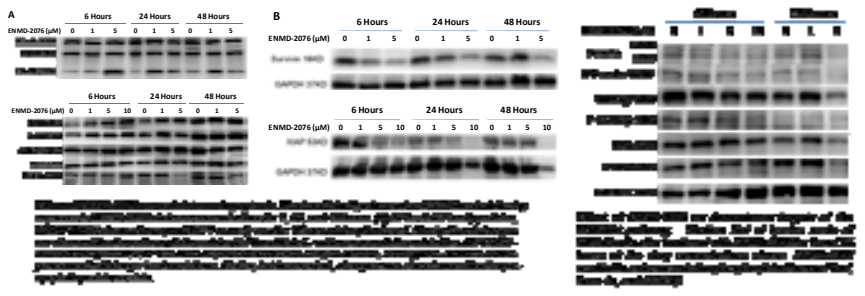
6 Hours
24 Hours
48 Hours
ENMD-2076 (µM)

4. ENMD-2076 inhibits aurora kinases A and B, and induces G2/M cell cycle arrest in MM cells.

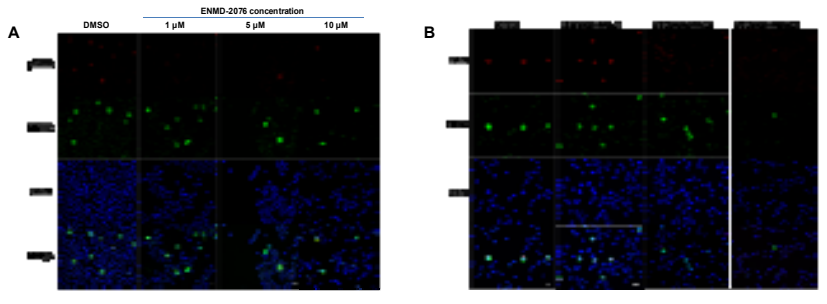
ENMD-2076 concentration
DMSO
1 µM
5 µM
10 µM

Immunofluorescence micrograph of MM1.R cells treated with ENMD-2076 for 24 hours, showing inhibition of (A) aurora A, and (B) aurora B. *pT288 reflects staining (red) for phospho-aurora A in panel A, p-H3 reflects staining of p-H3 Ser^{10} in panel B, MPM2 reflect staining of cells in mitosis, and DAPI staining of DNA is a general marker of cells. The bottom row shows merging of the images of each of the stains to assist in localizing of the stain to particular cells. As shown, ENMD-2076 resulted in a reduction of pT288 staining cells, indicating inhibition of aurora A.*

RESULTS

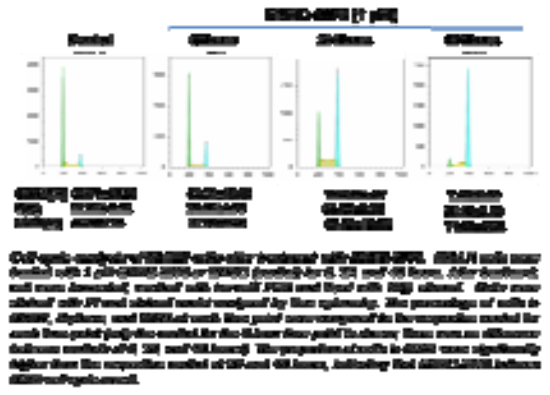

5. ENMD-2076 is active against MM cells *in vivo*.

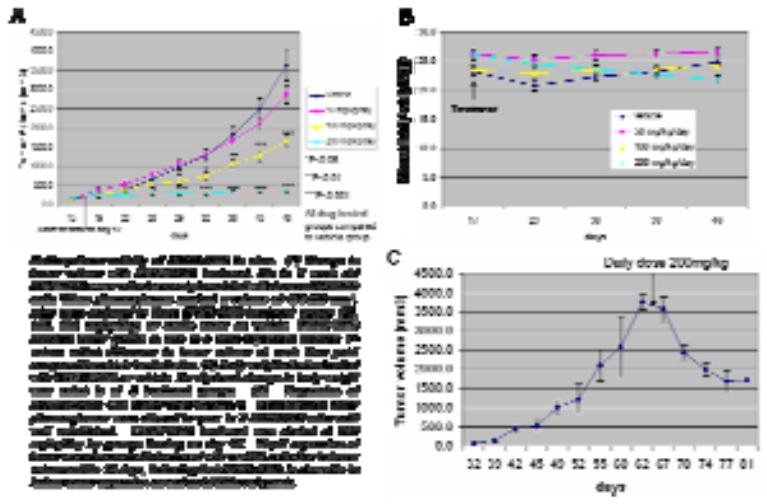
Daily dose 200mg/kg
days

6. ENMD-2076 inhibits proliferation, aurora kinases, ectopic FGFR-3 tyrosine kinase activity, and angiogenesis in MM cells *in vivo*.

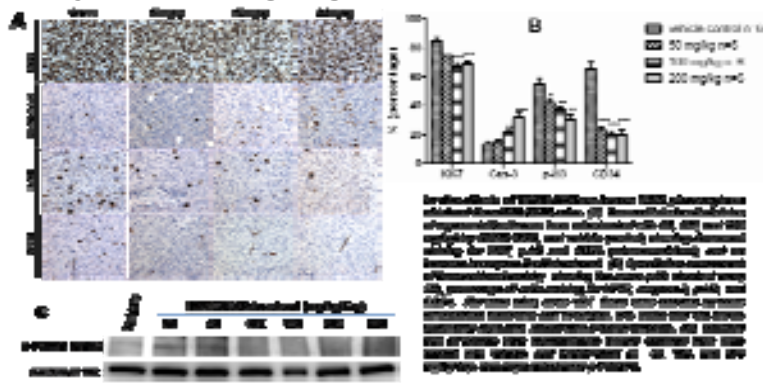

7. ENMD-2076 induces minimal toxicity to hematopoietic progenitors *in vitro* and *in vivo*.

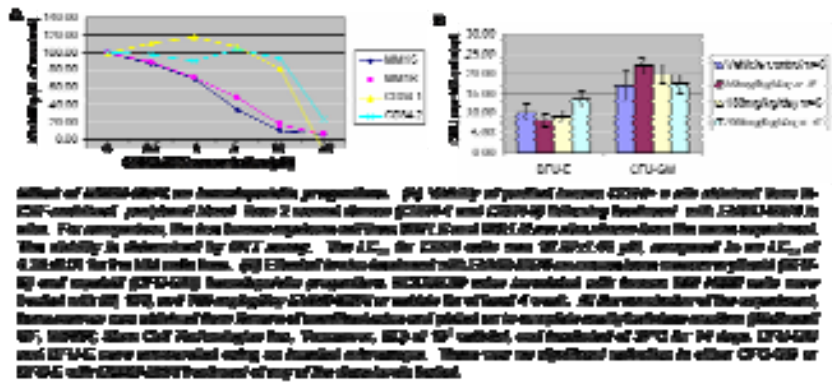

RESULTS

8. ENMD-2076 shows synergistic cytotoxic activity against MM cell lines when combined with lenalidomide.

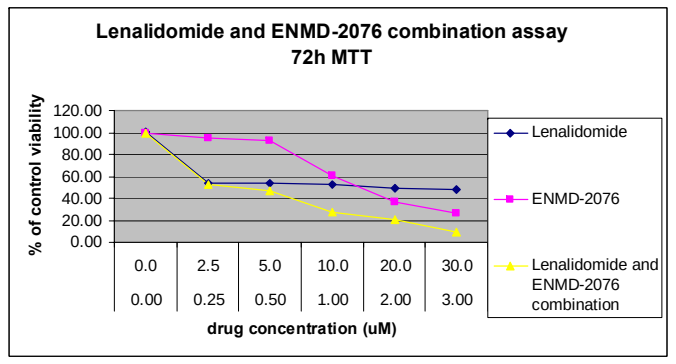
Lenalidomide and ENMD-2076 combination assay
72h MTT
% of control viability
120.00
100.00
80.00
60.00
40.00
20.00
0.00
0.0
2.5
5.0
10.0
20.0
30.0
0.00
0.25
0.50
1.00
2.00
3.00
drug concentration (uM)
Lenalidomide
ENMD-2076
Lenalidomide and ENMD-2076 combination

Using the Chou-Talalay method, the combination indices (CI) were < 1 for all three cell lines across a range of concentrations of ENMD-2076 free base (0.25-1.0 µM) plus lenalidomide (2.5-10µM) indicating synergistic activity.

Table 1. CIs generated from isobologram at different concentration of ENMD-2076 and lenalidomide

Cell line	ENMD-2076 (µM)	lenalidomide (µM)	Fractional activity	CI
H929	1	10	0.7276	0.362
MM1R	1	10	0.7455	0.315
MM1S	1	10	0.7184	0.404

CONCLUSIONS

1. ENMD-2076 alone shows significant *in vitro* and *in vivo* antitumor activity against MM cells, and acts via multiple mechanisms.
2. ENMD-2076 induces minimal cytotoxicity to human and murine hematopoietic progenitors *in vitro* and *in vivo*.
3. ENMD-2076 shows synergistic cytotoxic activity when combined with lenalidomide against MM cells.
4. The results provide rationale for the investigation of ENMD-2076 alone and in combination with lenalidomide in patients with multiple myeloma.